15328

Nalbuphine Oral Extended-Release Tablets Reduce Cough Bouts in Patients With Idiopathic Pulmonary Fibrosis

Jaclyn A. Smith,[1] Kevin J. Carroll,[2] David Clark,[3] Philip L. Molyneaux[4]

[1]The University of Manchester, Manchester, United Kingdom; [2]KJC Statistics Ltd, Woodford, United Kingdom; [3]Trevi Therapeutics, New Haven, CT, USA; [4]National Heart and Lung Institute, Imperial College London, London, United Kingdom

Background

- There are no approved therapies for cough for patients with idiopathic pulmonary fibrosis (IPF)[1]
- NAL ER is an oral extended release tablet that contains nalbuphine, a neuromodulator with a dual mechanism of action acting as a κ-opioid receptor agonist and a μ-opioid receptor antagonist[2]
- Nalbuphine acts centrally in the brain and peripherally in the lungs via the κ and μ receptors, which are important mediators of cough[3]
- In a randomized, double-blind, placebo-controlled, phase 2a crossover trial (CANAL; NCT04030026), patients with IPF who were treated with oral NAL ER had a 76.1% reduction in 24-hour cough frequency compared with a 25.3% reduction in those receiving placebo[4]
- Coughs typically cluster within bouts defined as clusters of sequential coughs, which are thought to contribute to the severity of cough[5]

Objective

- This post hoc analysis of the CANAL trial was conducted to examine the effect of NAL ER on cough bouts in patients with IPF

Methods

- Participants aged ≥18 years who had an IPF diagnosis, self-reported chronic cough lasting ≥8 weeks, and daytime cough severity score ≥4 on the Cough Severity Numerical Rating Scale (CS-NRS) were enrolled in the CANAL trial[4] (**Figure 1**)
- In the CANAL crossover trial, patients were randomly assigned 1:1 to receive either NAL ER during treatment period 1 followed by placebo in treatment period 2 or placebo during treatment period 1 followed by NAL ER in treatment period 2 (**Figure 1**)
- 24-hour cough frequency data from a digital cough monitor (VitaloJAK®, Vitalograph®) at day −1 (baseline) and day 21 (end of treatment) of the CANAL trial were used to analyze cough bouts
- Using a validated custom-written algorithm, cough bouts (a cough frequency consisting of 2 or more coughs) were generated by combining consecutive cough sounds within 1 second, 2 seconds, 3 seconds, 5 seconds, and 10 seconds of one another
- Data were analyzed using the negative binomial model and mixed-model for repeated-measures analysis

Results

- Cough data were analyzed from 38 patients (n=19, per arm) (**Figure 1**)

Figure 1. Study Design

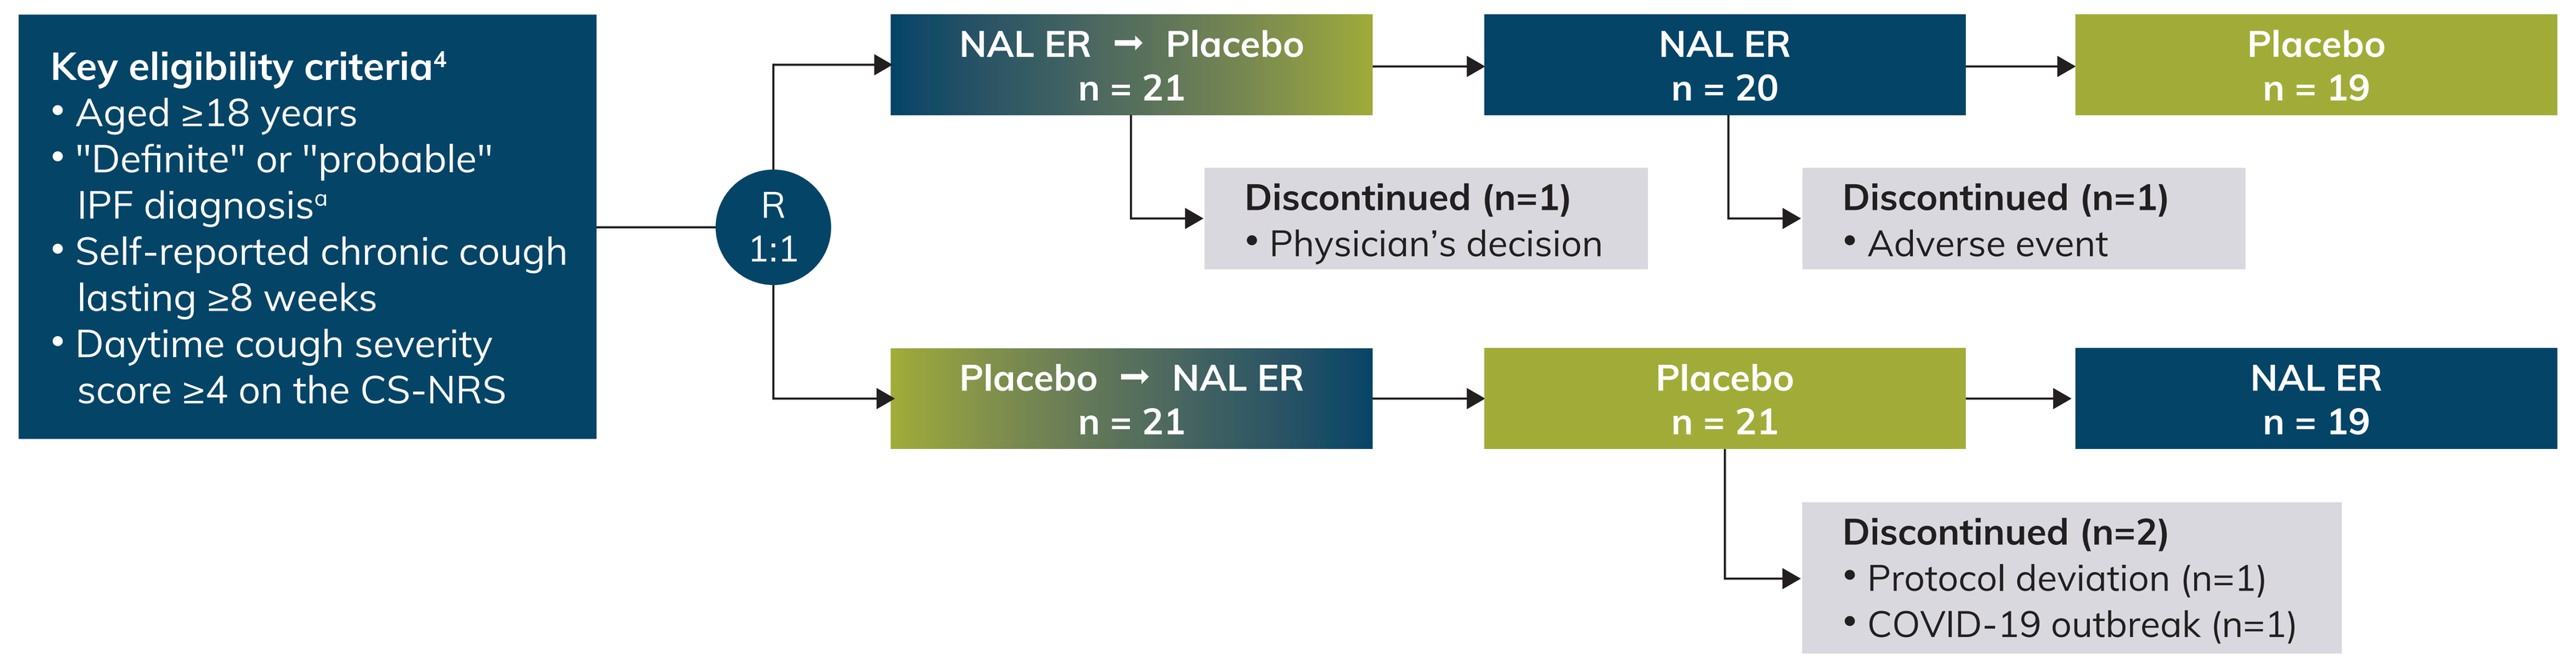
Key eligibility criteria[4]
• Aged ≥18 years
• "Definite" or "probable" IPF diagnosis[a]
• Self-reported chronic cough lasting ≥8 weeks
• Daytime cough severity score ≥4 on the CS-NRS
R 1:1
NAL ER → Placebo n = 21
NAL ER n = 20
Placebo n = 19
Discontinued (n=1) • Physician's decision
Discontinued (n=1) • Adverse event
Placebo → NAL ER n = 21
Placebo n = 21
NAL ER n = 19
Discontinued (n=2) • Protocol deviation (n=1) • COVID-19 outbreak (n=1)

[a]In accordance with international guidelines current at the time of recruitment.[5]

- Baseline cough bout rates were similar between both arms before treatment using negative binominal analysis (**Figure 2**)

Figure 2. Baseline Mean Cough Bout Rate (95% CI) Using Negative Binominal Analysis

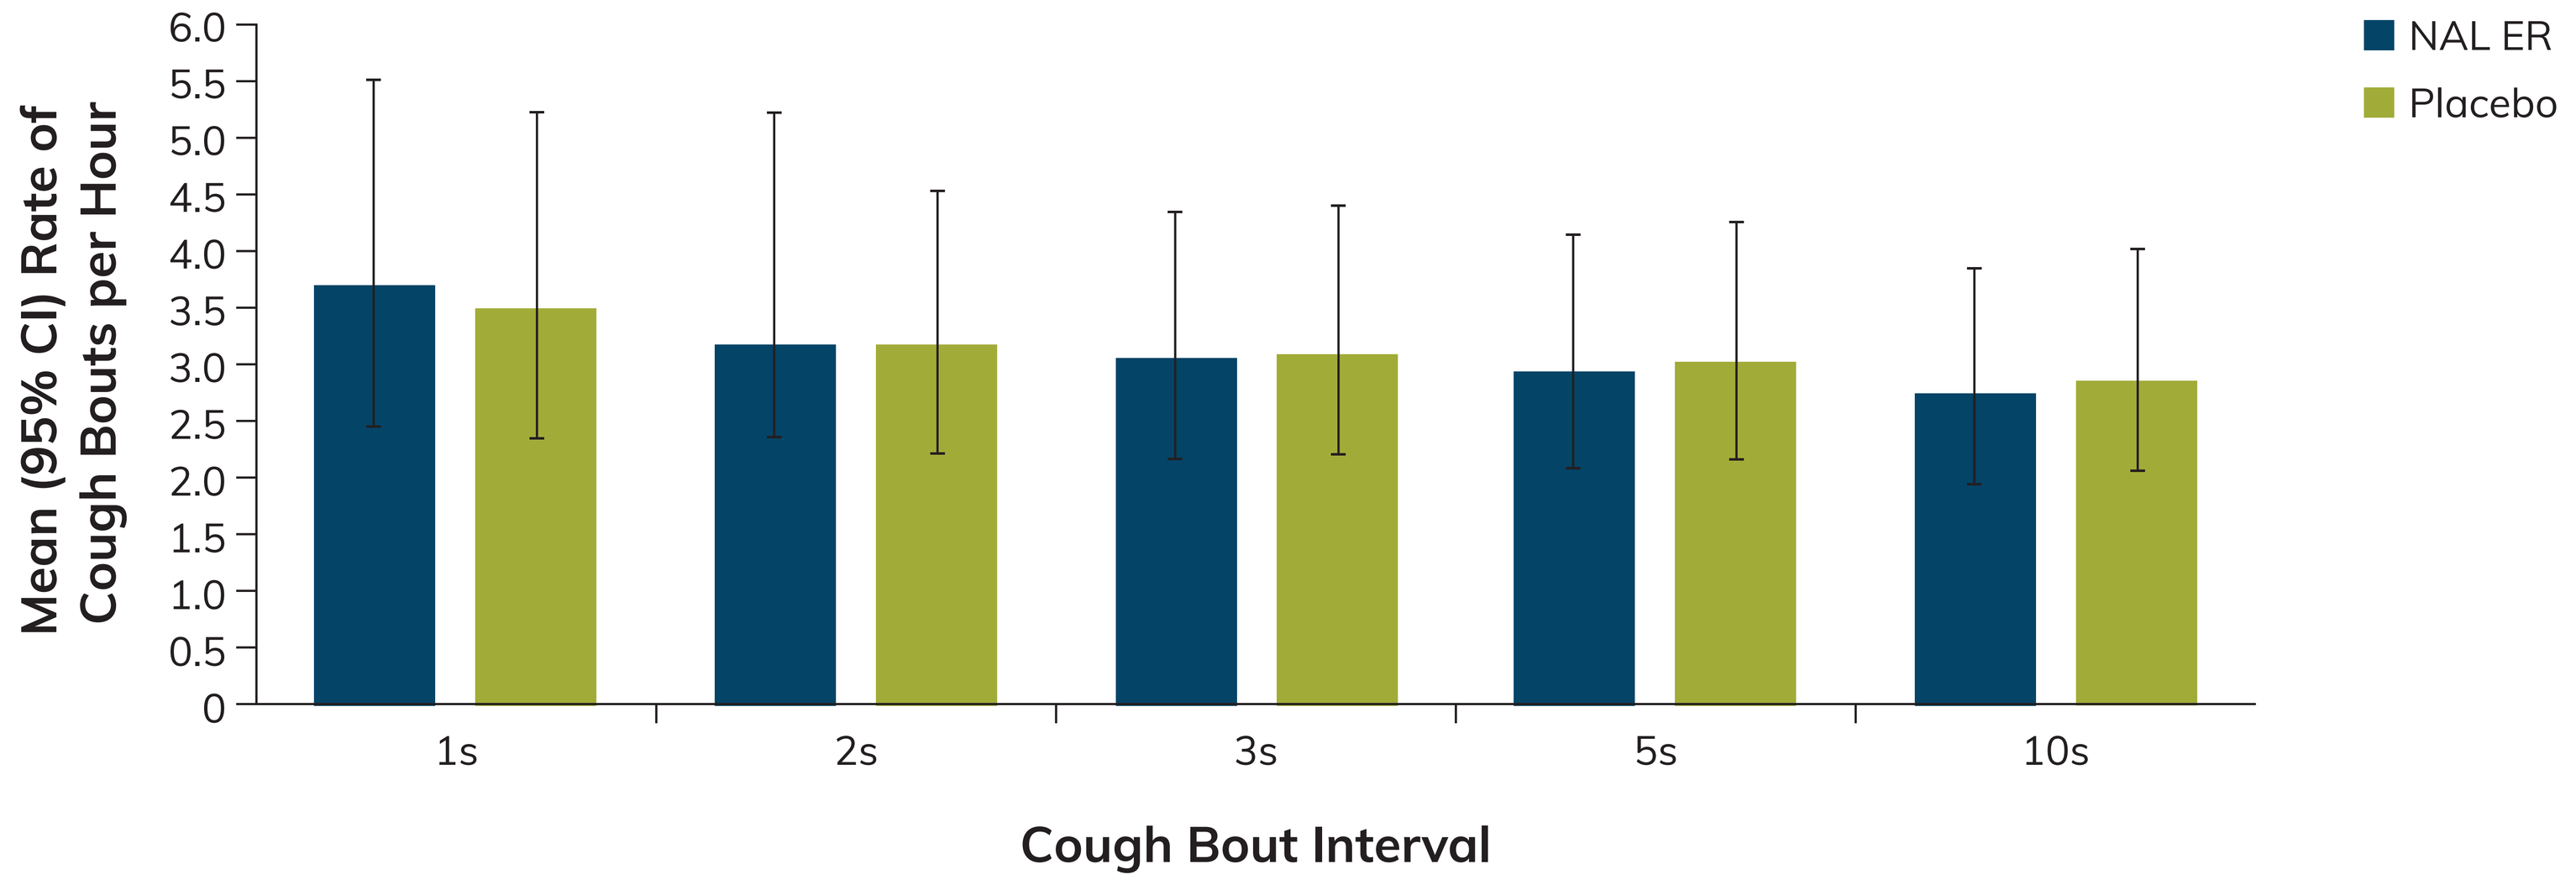
Mean (95% CI) Rate of Cough Bouts per Hour
6.0
5.5
5.0
4.5
4.0
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0
1s
2s
3s
5s
10s
Cough Bout Interval
NAL ER
Placebo

Cough bout is defined as a sequence of 2 or more coughs.

- Compared with placebo, at day 21, treatment with NAL ER reduced the mean rate of cough bouts using negative binominal analysis, regardless of the interval of time used to define a cough bout (**Figure 3**)

Figure 3. Mean Cough Bout Rate (95% CI) Using Negative Binominal Analysis at Day 21

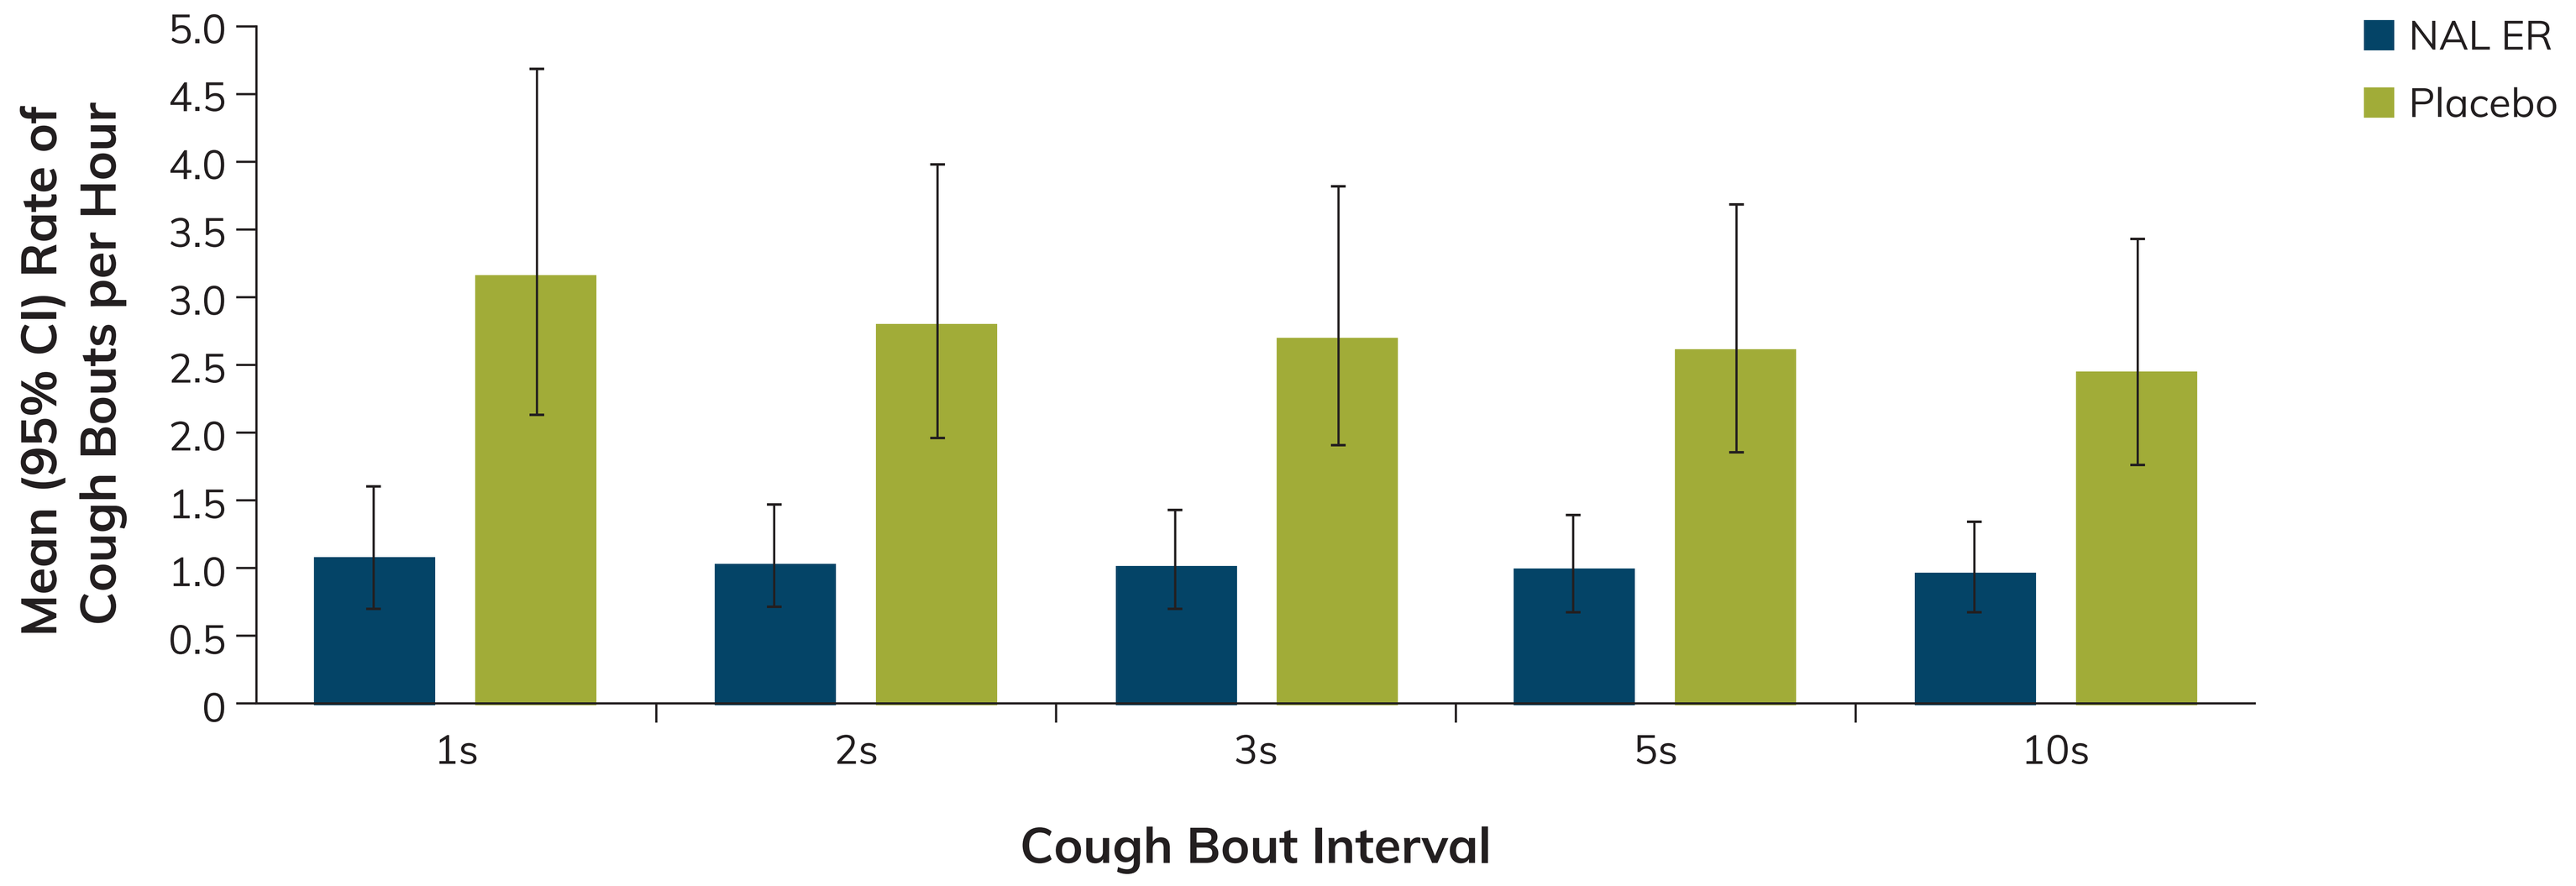
Mean (95% CI) Rate of Cough Bouts per Hour
5.0
4.5
4.0
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0
1s
2s
3s
5s
10s
Cough Bout Interval
NAL ER
Placebo

Cough bout is defined as a sequence of 2 or more coughs.

- The percentage change from baseline in mean cough bout rates was significantly reduced in patients treated with NAL ER compared with placebo (**Figure 4**)
 - The reduction from baseline in the NAL ER group was −68.4% to −73.2% versus <20% with placebo (all *P* < .0001; **Figure 4**)
 - These results were driven by consistently large and highly significant reductions from baseline in cough bout rate with NAL ER regardless of cough bout definition
 - Corresponding reductions with placebo were considerably smaller in magnitude

Figure 4. Percentage Change From Baseline (95% CI) in Mean Cough Bout Rate Using Negative Binominal Analysis at Day 21

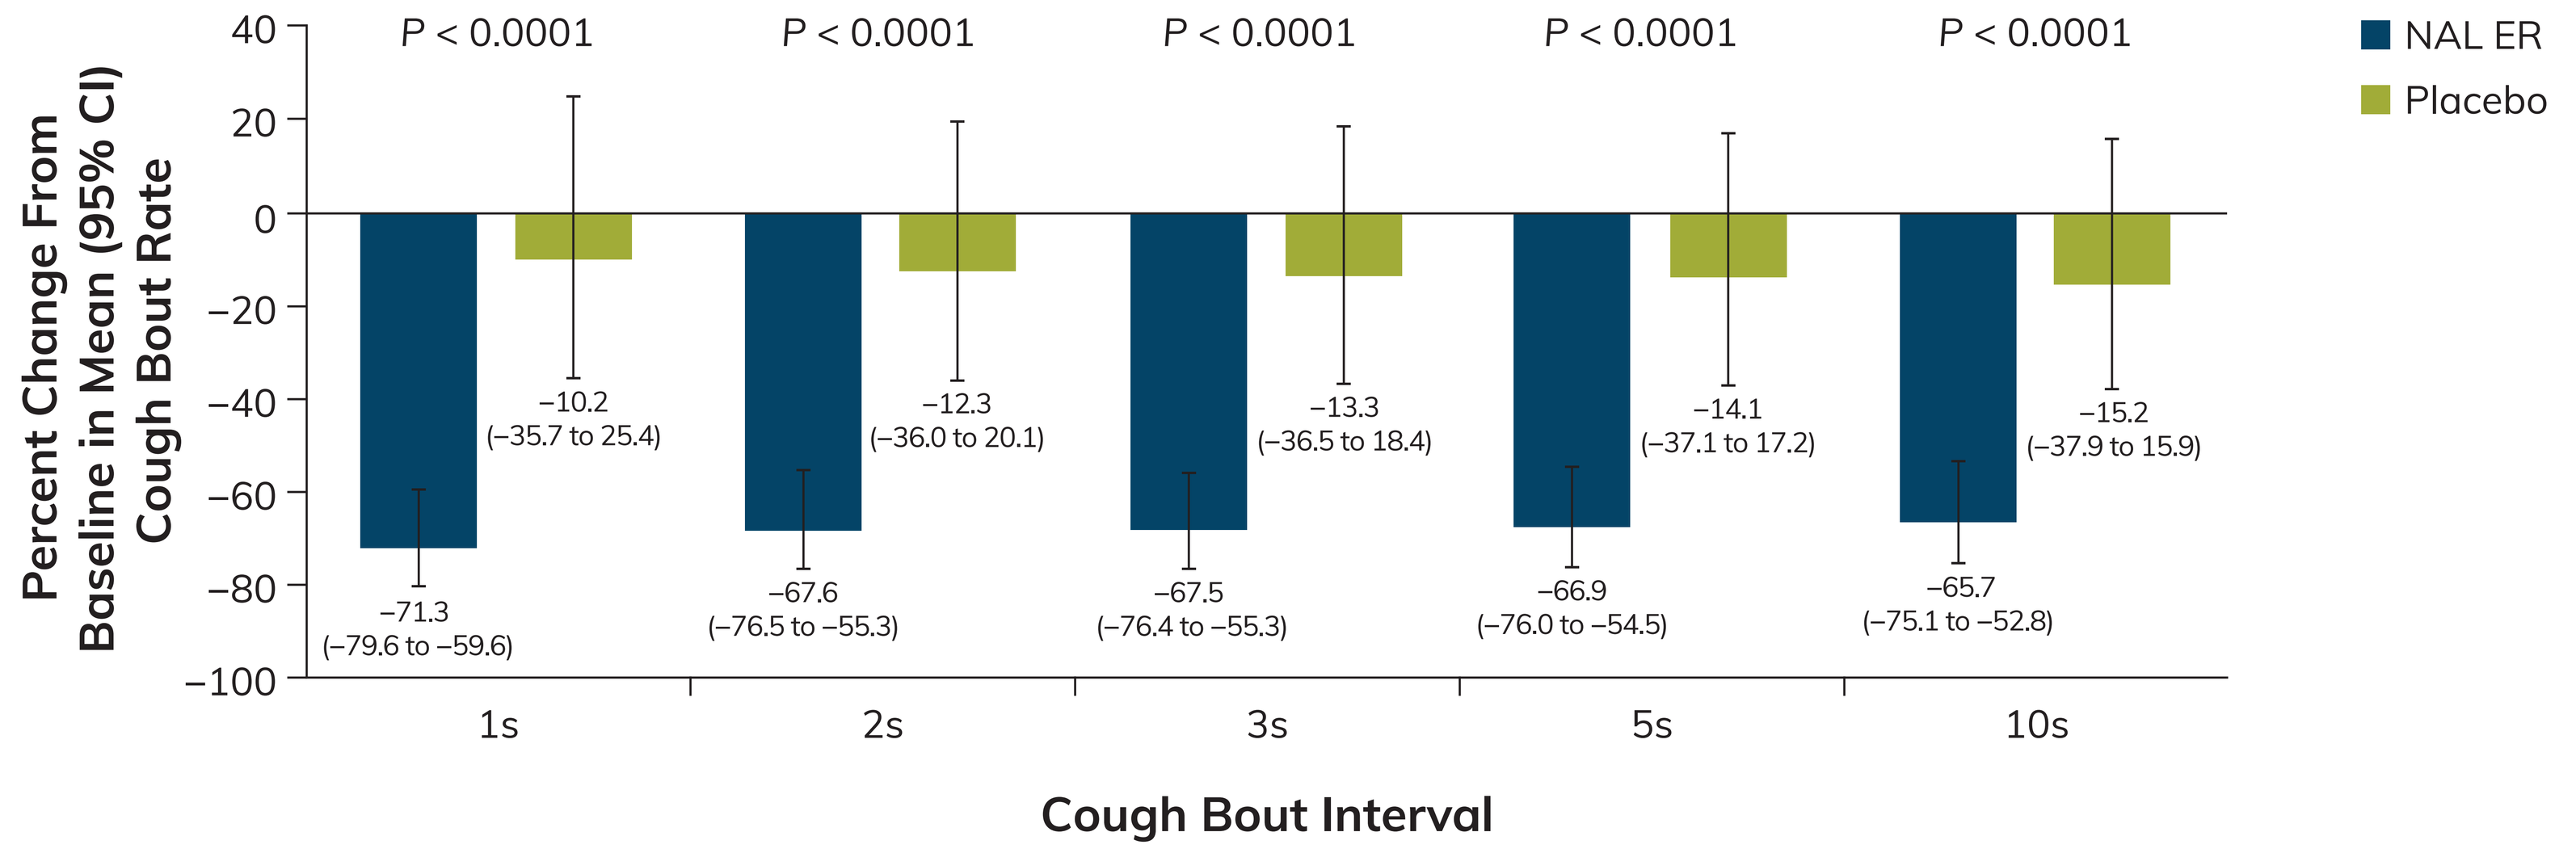
Percent Change From Baseline in Mean (95% CI) Cough Bout Rate
40
20
0
−20
−40
−60
−80
−100
P < 0.0001
P < 0.0001
P < 0.0001
P < 0.0001
P < 0.0001
−71.3 (−79.6 to −59.6)
−10.2 (−35.7 to 25.4)
−67.6 (−76.5 to −55.3)
−12.3 (−36.0 to 20.1)
−67.5 (−76.4 to −55.3)
−13.3 (−36.5 to 18.4)
−66.9 (−76.0 to −54.5)
−14.1 (−37.1 to 17.2)
−65.7 (−75.1 to −52.8)
−15.2 (−37.9 to 15.9)
1s
2s
3s
5s
10s
Cough Bout Interval
NAL ER
Placebo

Cough bout is defined as a cough frequency of more than 2 coughs.

- Results were consistent when analyzing log-transformed data via a mixed model for repeated measures (**Figure 5**)

Figure 5. Geometric Mean Cough Bout Rate per Hour (95% CI) Using Mixed Model for Repeated Measures at Day 21

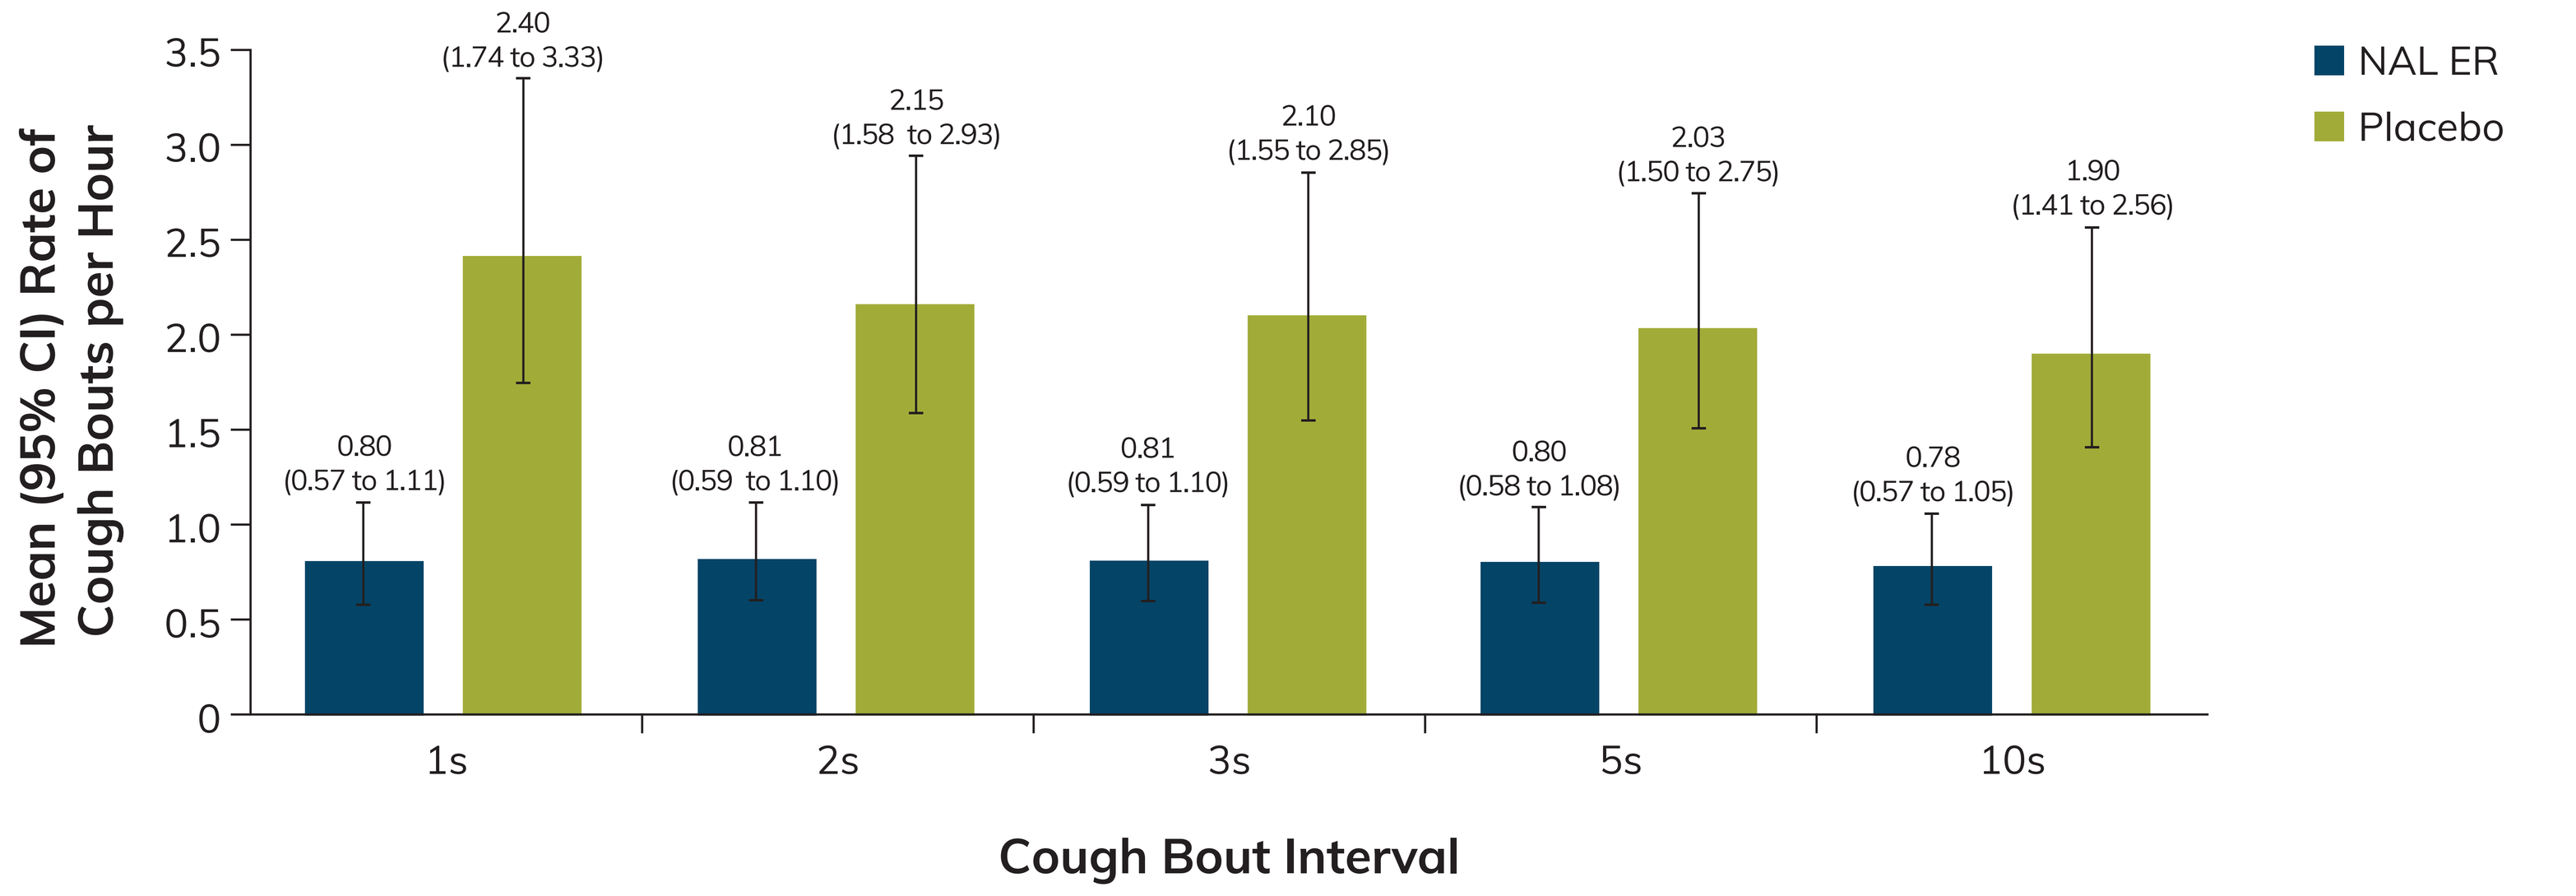
Mean (95% CI) Rate of Cough Bouts per Hour
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0
0.80 (0.57 to 1.11)
2.40 (1.74 to 3.33)
0.81 (0.59 to 1.10)
2.15 (1.58 to 2.93)
0.81 (0.59 to 1.10)
2.10 (1.55 to 2.85)
0.80 (0.58 to 1.08)
2.03 (1.50 to 2.75)
0.78 (0.57 to 1.05)
1.90 (1.41 to 2.56)
1s
2s
3s
5s
10s
Cough Bout Interval
NAL ER
Placebo

Cough bout is defined as a cough frequency of more than 2 coughs.

- Percentage change from baseline in mean cough bout rate via a log-transformed mixed-model for repeated-measures analysis showed a significant reduction (all *P* < .0001) from baseline in the mean rate of cough bouts with NAL ER compared with placebo (**Figure 6**)
 - Reductions of approximately 75% were observed with use of NAL ER compared with <20% with use of placebo (**Figure 6**)

Figure 6. Percentage Change From Baseline (95% CI) in Mean Cough Bout Rate Using Mixed Model for Repeated Measures at Day 21

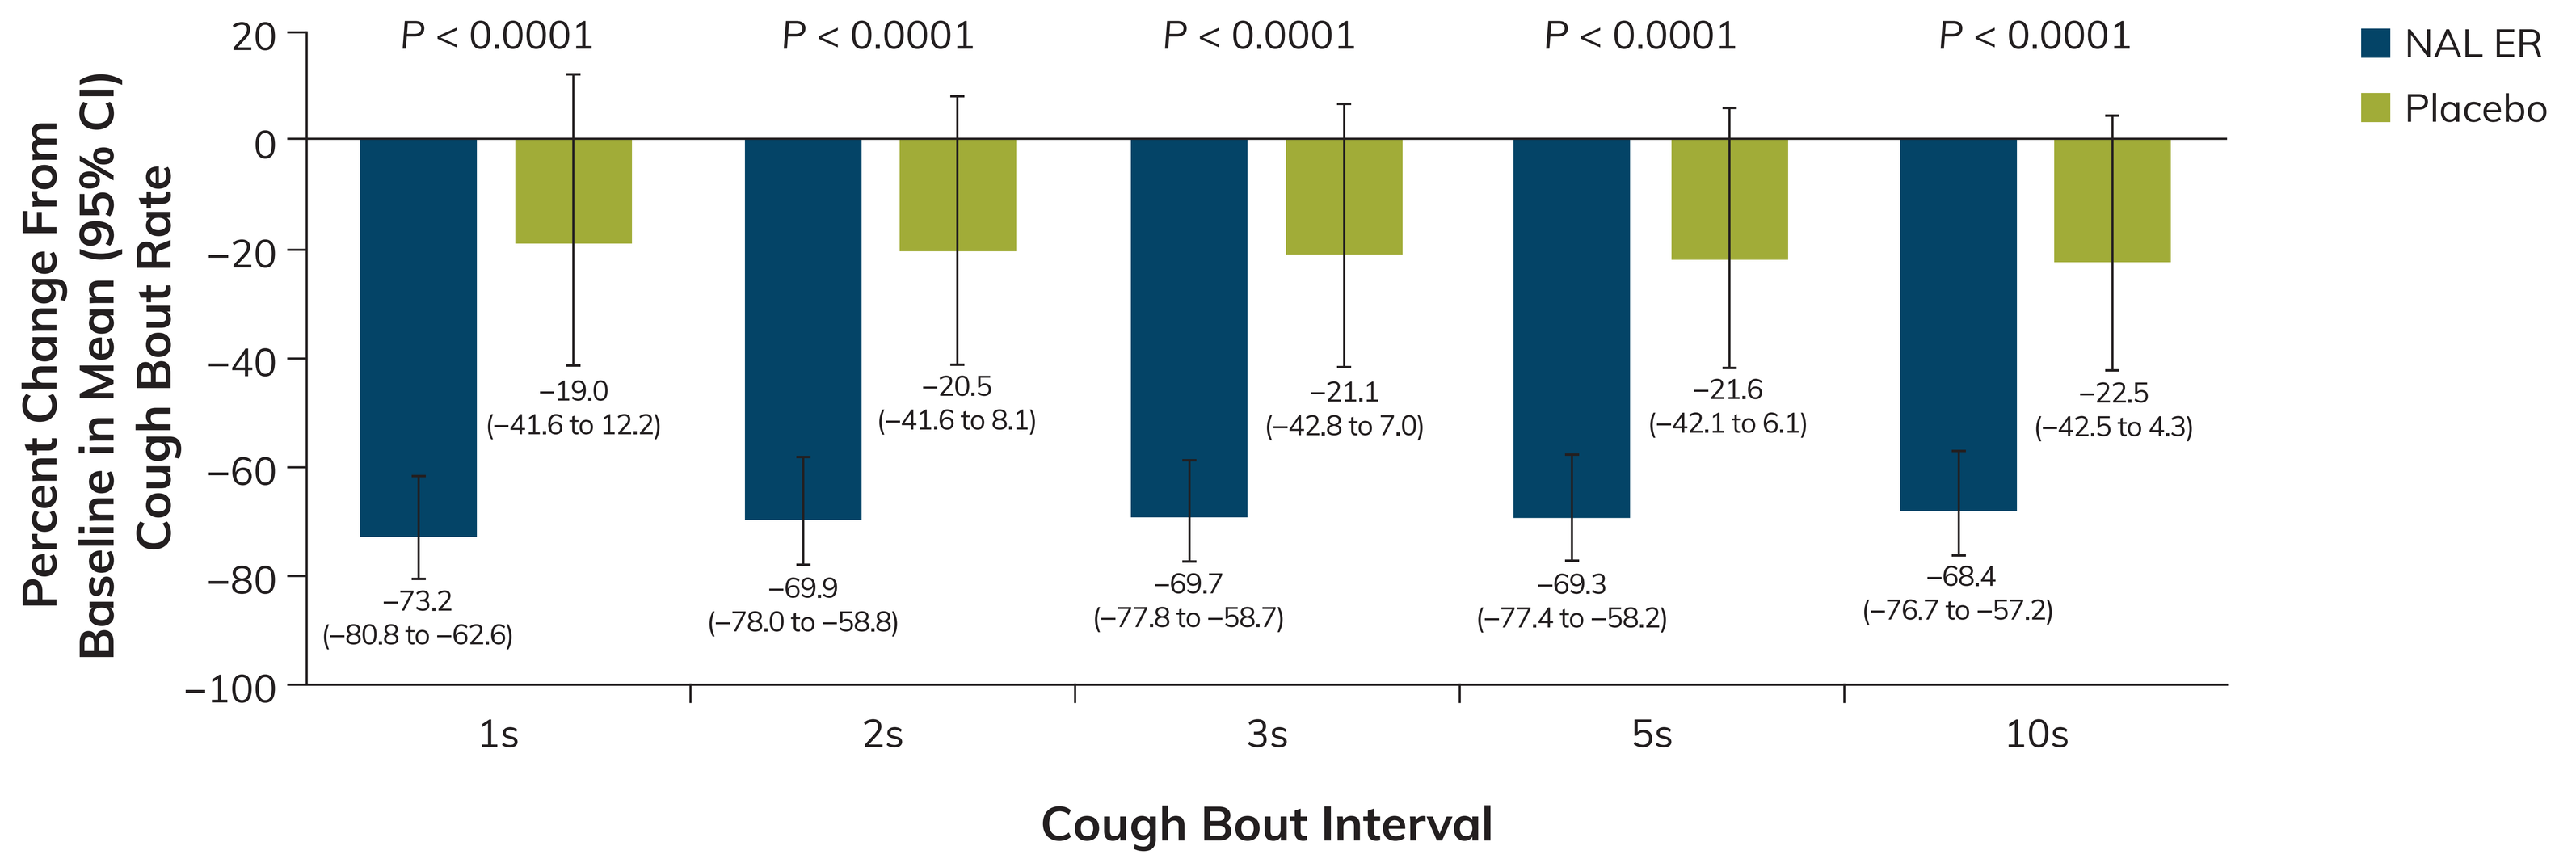
Percent Change From Baseline in Mean (95% CI) Cough Bout Rate
20
0
−20
−40
−60
−80
−100
P < 0.0001
P < 0.0001
P < 0.0001
P < 0.0001
P < 0.0001
−73.2 (−80.8 to −62.6)
−19.0 (−41.6 to 12.2)
−69.9 (−78.0 to −58.8)
−20.5 (−41.6 to 8.1)
−69.7 (−77.8 to −58.7)
−21.1 (−42.8 to 7.0)
−69.3 (−77.4 to −58.2)
−21.6 (−42.1 to 6.1)
−68.4 (−76.7 to −57.2)
−22.5 (−42.5 to 4.3)
1s
2s
3s
5s
10s
Cough Bout Interval
NAL ER
Placebo

Cough bout is defined as a cough frequency of more than 2 coughs.

Conclusions

- Consistent with results observed in the primary end point analysis in the CANAL trial,[3] use of NAL ER significantly reduced both the mean number and the rate of cough bouts compared with placebo in patients with IPF, regardless of the length of time used to define a cough bout and using different statistical analyses
- NAL ER is the first drug to show significant reduction in cough bouts in patients with IPF, and this effect may be greater than the effect on standard cough frequency

References

1. Vigeland CL et al. *Respir Med.* 2017;123:98-104.
2. Hawi A et al. *BMC Nephrol.* 2015;16:47.
3. Adcock JJ. *Respir Med.* 1991;85(suppl A):43-46.
4. Maher TM et al. *NEJM Evid.* 2023;2(8):EVIDoa2300083.
5. Lee KK et al. *Chest.* 2021;159(1):282-293.
6. Raghu G et al. *Am J Respir Crit Care Med.* 2018;198(5):e44-e68.

Abbreviations

CI, confidence interval; COVID-19, coronavirus disease 2019; CS-NRS, Cough Severity Numerical Rating Scale; IPF, idiopathic pulmonary fibrosis; NAL ER, nalbuphine extended-release; s, second.

Acknowledgments

The authors thank the investigators of the CANAL post hoc study. The sponsor of the study and the investigators thank the patients and their families for their participation in and support of this study. This study was sponsored by Trevi Therapeutics (New Haven, CT, USA). Medical writing assistance was provided by ApotheCom (San Francisco, CA, USA) and funded by Trevi Therapeutics.

Disclosures

Jaclyn Smith discloses consultancy work for Trevi Therapeutics and a hospital license agreement with Vitalograph for the use of cough detection software. The hospital receives royalties, which may be distributed to the department with which Jaclyn Smith is affiliated.

Presented at the
American Thoracic Society International Conference;
May 17-22, 2024; San Diego, California