Utility of EXACT, a patient-reported outcome instrument, for idiopathic pulmonary fibrosis-related cough

Marlies Wijsenbeek,[1] Thomas Sciascia,[2] Toby M Maher,[3,4] Philip L Molyneaux[4]

[1]Department of Respiratory Medicine, Erasmus Medical Center, Rotterdam, Netherlands; [2]Trevi Therapeutics, New Haven, CT, USA; [3]Keck School of Medicine, University of Southern California, Los Angeles, CA, USA; [4]National Heart and Lung Institute, Imperial College London, London, UK

BACKGROUND

- Cough is common in individuals with idiopathic pulmonary fibrosis (IPF), contributing to disease-associated morbidity and diminished quality of life through effects on breathlessness, and an individual's ability to work and socialize.[1]
- Practical, validated patient diaries are needed to evaluate idiopathic pulmonary fibrosis respiratory symptoms.
- EXACT is a patient-reported outcome (PRO) developed for COPD.
 - Cough and breathlessness subdomains of the EXACT outcome have been used to evaluate patients with IPF, called the Evaluating Respiratory Symptoms (E-RS:IPF) tool.[2]
- Nalbuphine extended release (NAL ER) is a κ-receptor agonist and μ-receptor antagonist that has been investigated in patients with IPF in the CANAL trial (NCT04030026).[3]

OBJECTIVE

- To identify optimal patient-reported endpoints in IPF studies by reporting on EXACT total score vs. E-RS cough and breathlessness subdomain scores, and comparing them to the objective NAL ER efficacy data from the CANAL trial.

METHODS

- The CANAL trial, randomised, double-blind, placebo-controlled, crossover trial with two 22-day treatment periods separated by a 2-week washout period was conducted (**Figure 1**).
 - Adults diagnosed with definite/probable IPF using international criteria and chronic cough for >8 weeks were enrolled.
- EXACT total score throughout the study was a secondary endpoint of the CANAL trial and was compared with the following endpoints:
 - E-RS:IPF cough subdomain score.
 - E-RS:IPF breathlessness subdomain score throughout the study.
 - Primary endpoint: mean change in objective daytime cough frequency (digital cough recorder VitaloJAK®) at Day 22.

Figure 1. CANAL study design.

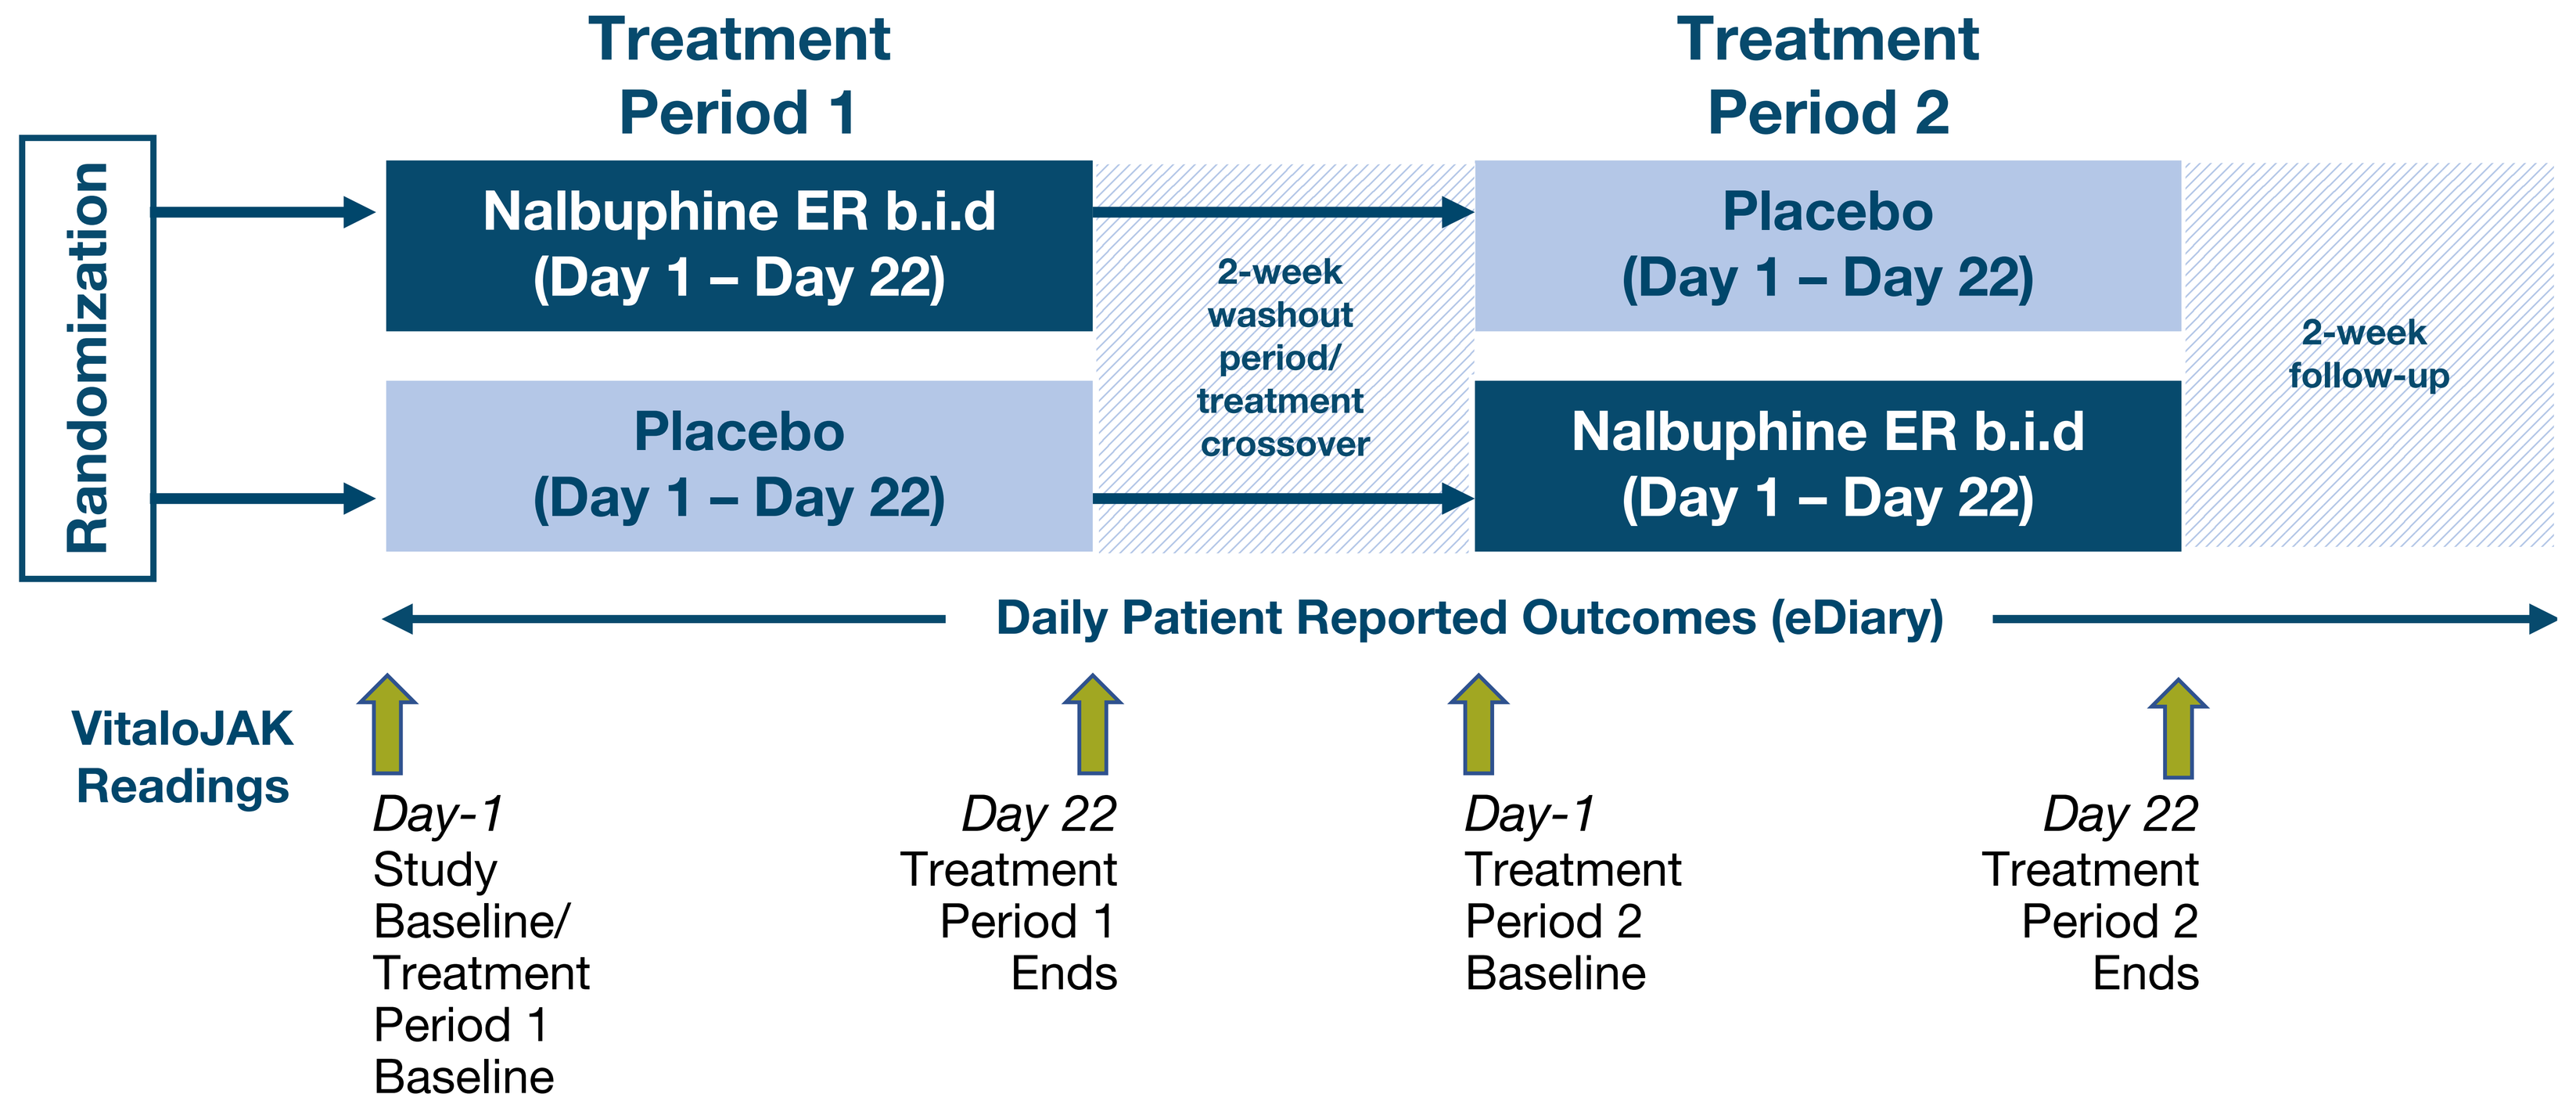
Treatment Period 1
Treatment Period 2
Randomization
Nalbuphine ER b.i.d (Day 1 – Day 22)
Placebo (Day 1 – Day 22)
2-week washout period/ treatment crossover
Placebo (Day 1 – Day 22)
Nalbuphine ER b.i.d (Day 1 – Day 22)
2-week follow-up
Daily Patient Reported Outcomes (eDiary)
VitaloJAK Readings
Day-1 Study Baseline/ Treatment Period 1 Baseline
Day 22 Treatment Period 1 Ends
Day-1 Treatment Period 2 Baseline
Day 22 Treatment Period 2 Ends

b.i.d., two times per day; NAL ER, nalbuphine extended release.

RESULTS

Figure 2. Mean change from baseline in EXACT overall score.

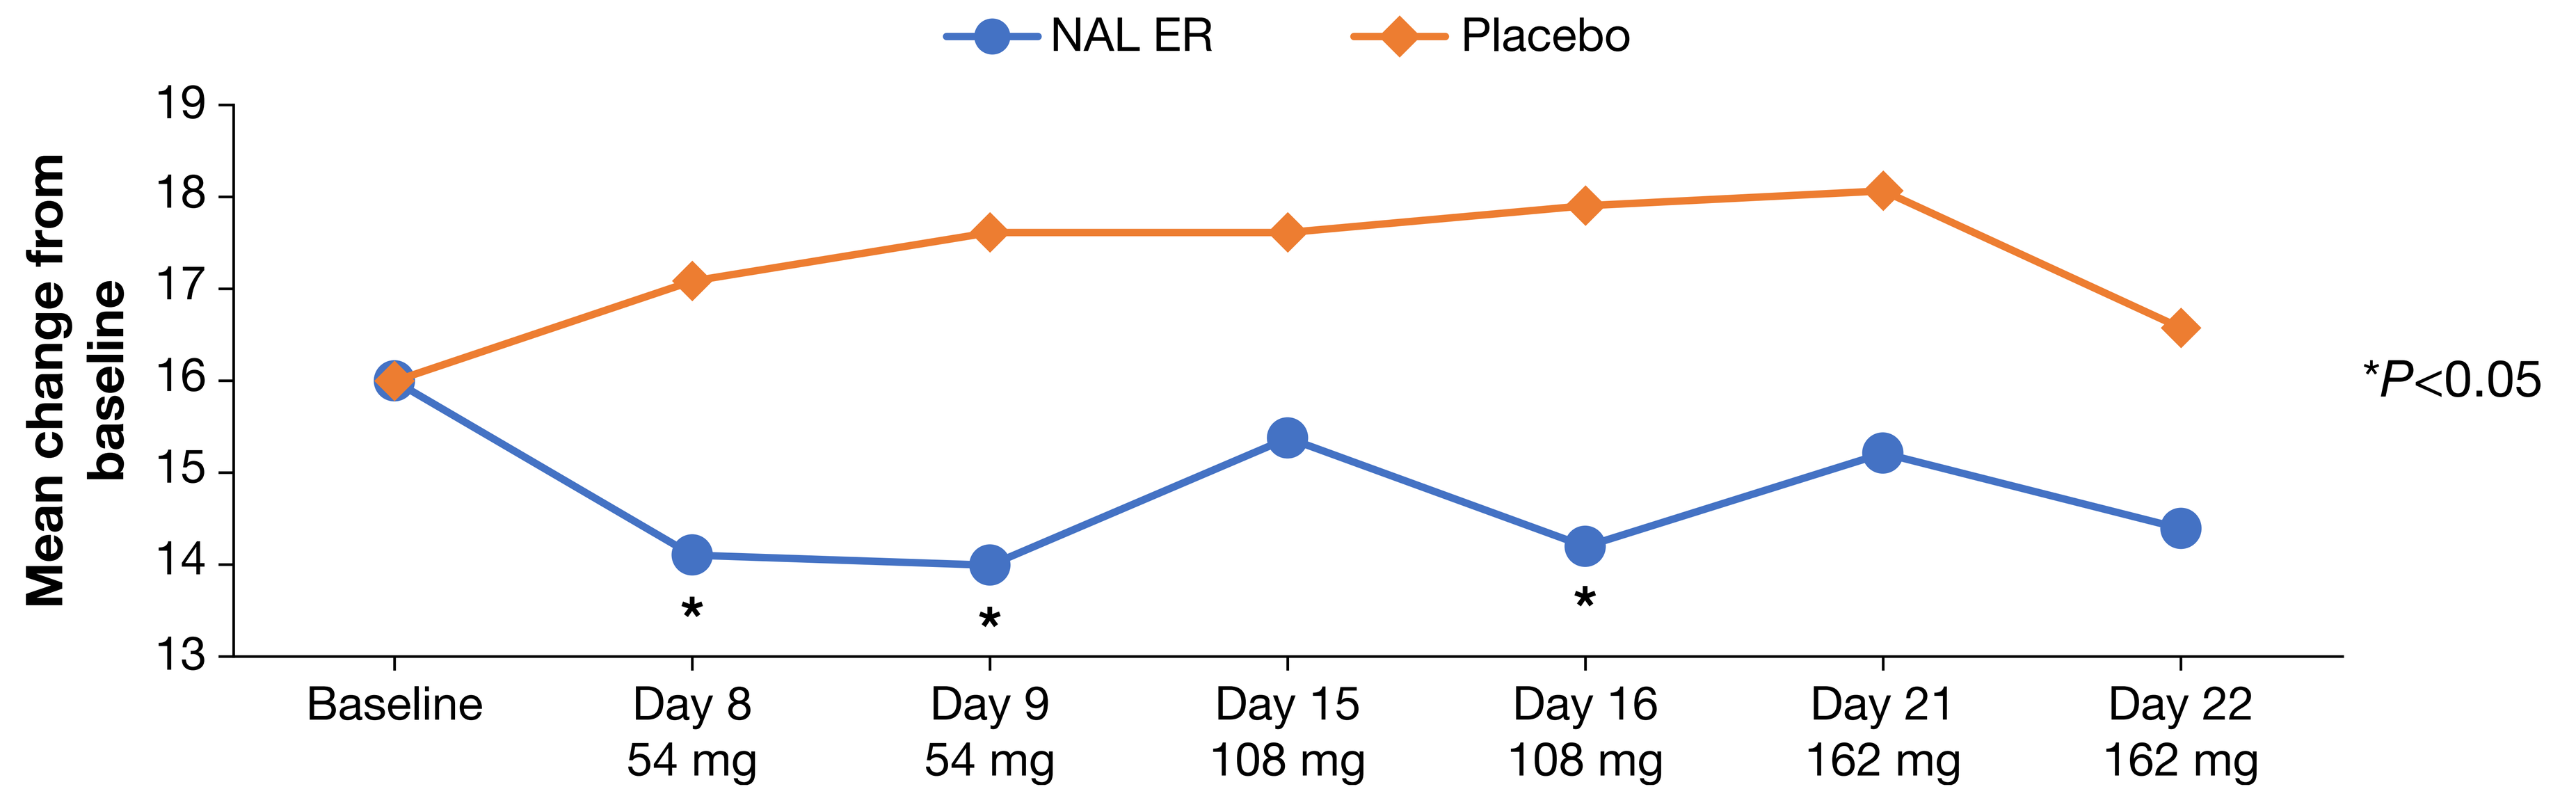
NAL ER
Placebo
Mean change from baseline
Baseline
Day 8 54 mg
Day 9 54 mg
Day 15 108 mg
Day 16 108 mg
Day 21 162 mg
Day 22 162 mg
*P<0.05

NAL ER, nalbuphine extended release.

- EXACT total score showed numerical improvements with NAL ER vs. placebo at all timepoints, reaching significance on Days 8, 9, and 16 (**Figure 2**).

Figure 3. Mean change from baseline in E-RS:IPF cough subdomain score.

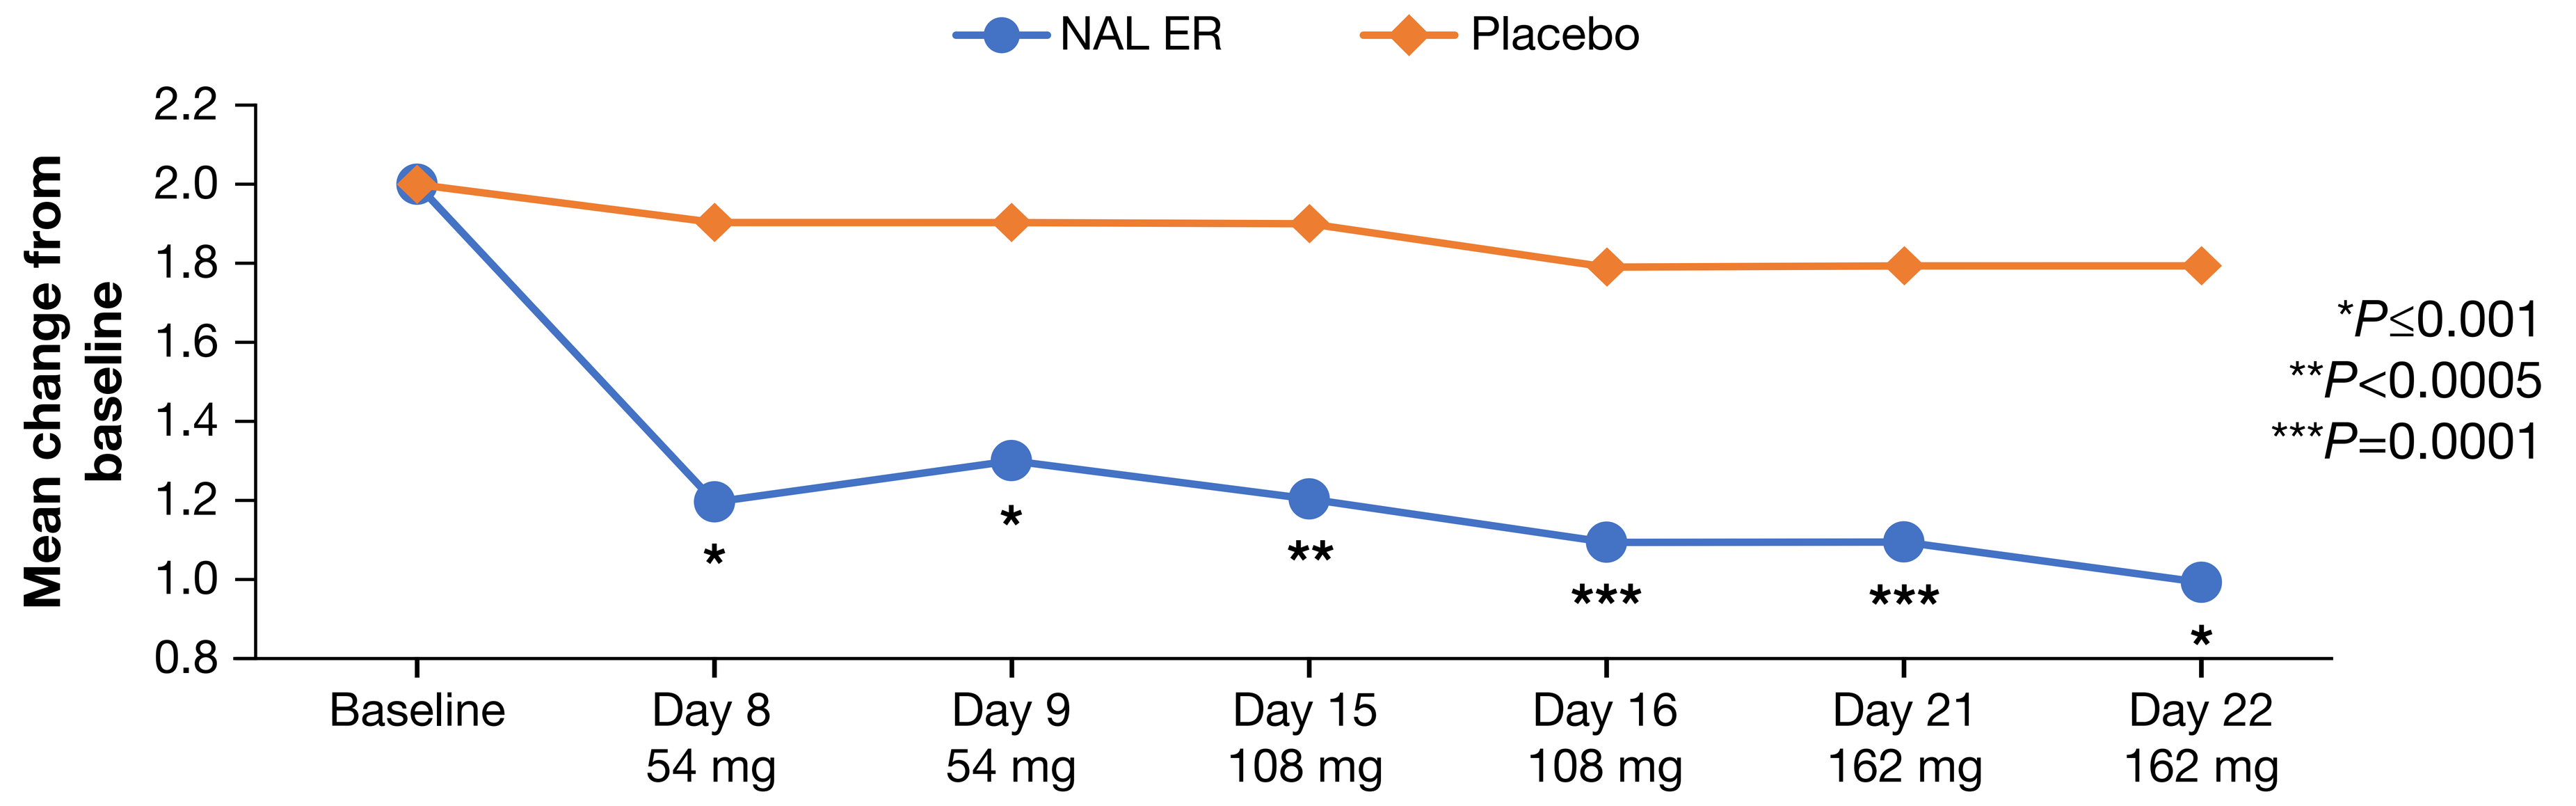
NAL ER
Placebo
Mean change from baseline
Baseline
Day 8 54 mg
Day 9 54 mg
Day 15 108 mg
Day 16 108 mg
Day 21 162 mg
Day 22 162 mg
*P≤0.001
**P<0.0005
***P=0.0001

E-RS, Evaluating Respiratory Symptoms subdomain; IPF, idiopathic pulmonary fibrosis; NAL ER, nalbuphine extended release.

- The mean changes from baseline in E-RS subdomain cough score were significantly improved for NAL ER vs. placebo at all timepoints (**Figure 3**).

Figure 4. Mean change from baseline in E-RS:IPF breathlessness subdomain score.

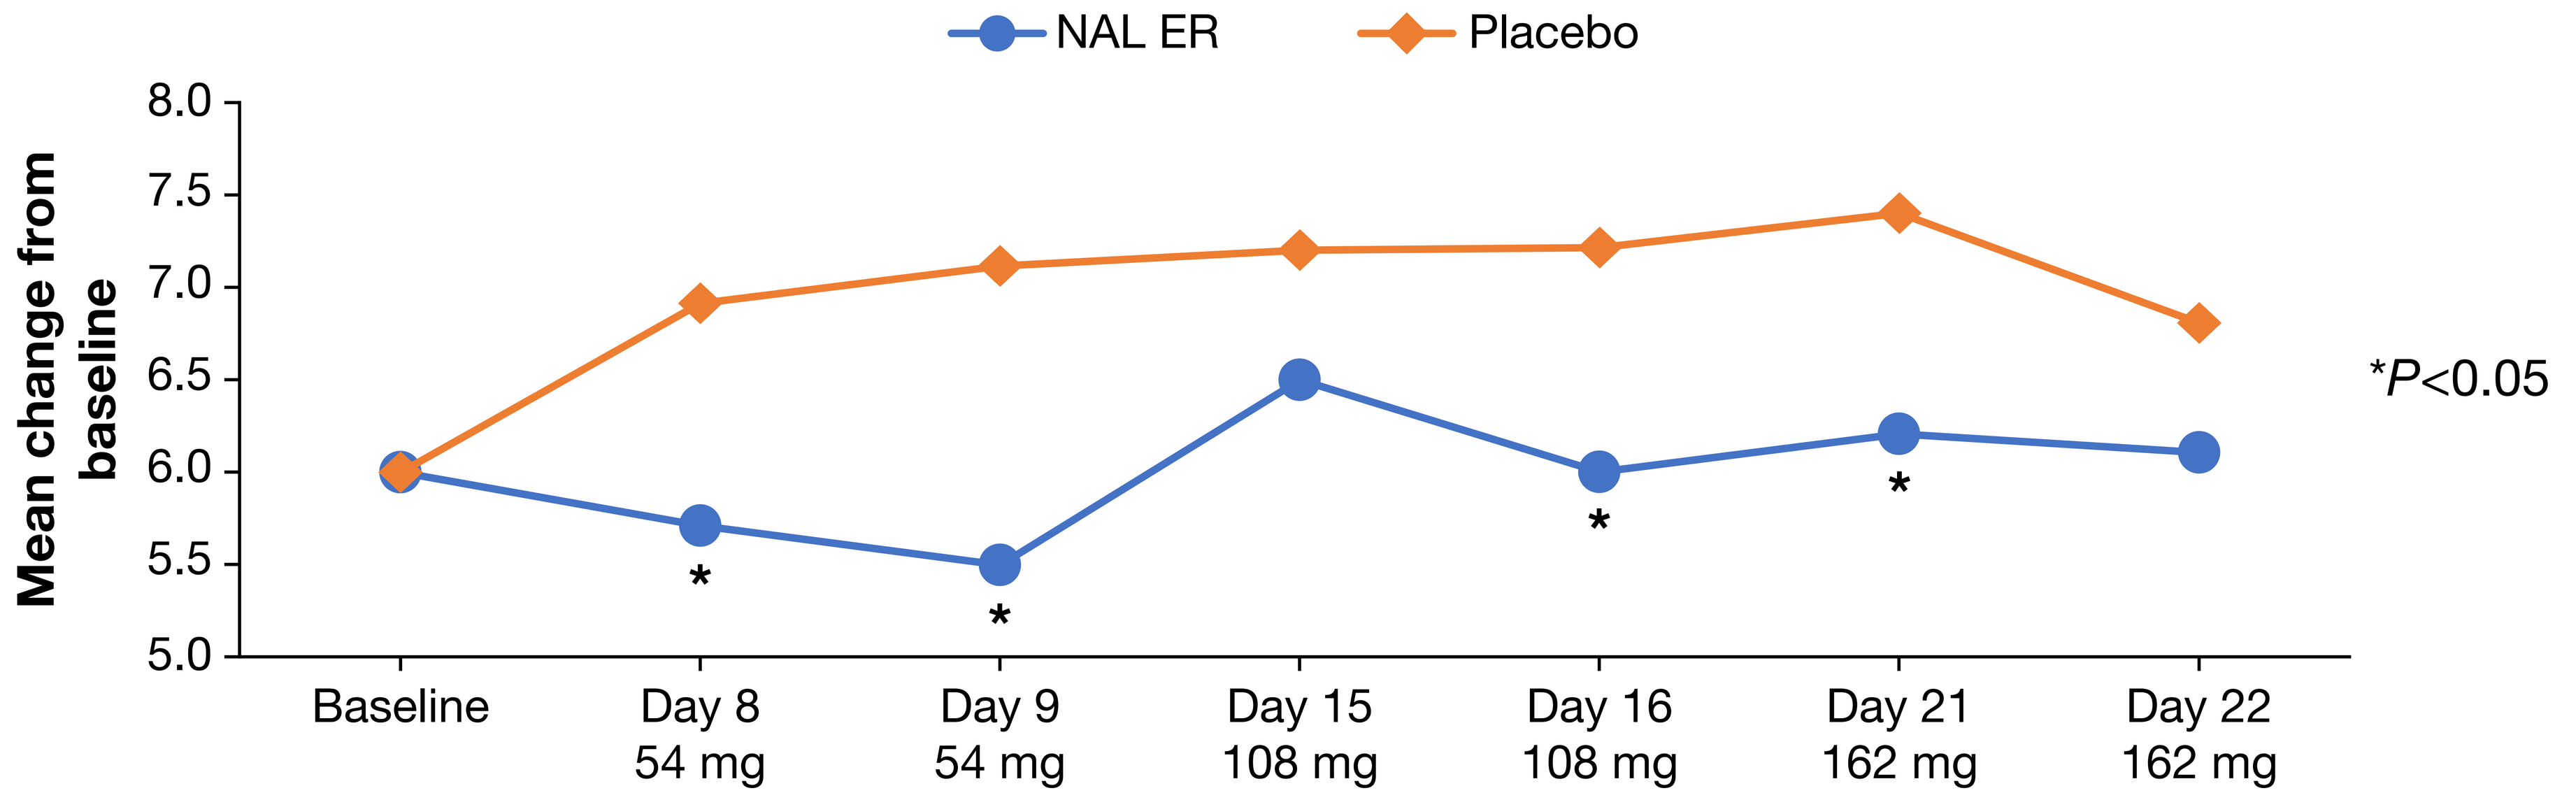
NAL ER
Placebo
Mean change from baseline
Baseline
Day 8 54 mg
Day 9 54 mg
Day 15 108 mg
Day 16 108 mg
Day 21 162 mg
Day 22 162 mg
*P<0.05

E-RS, Evaluating Respiratory Symptoms subdomain; IPF, idiopathic pulmonary fibrosis; NAL ER, nalbuphine extended release.

- Breathlessness subdomain scores showed similar trends to the EXACT total score: E-RS breathlessness scores for patients treated with NAL ER were numerically improved vs. placebo, reaching significance on Days 8, 9, 16, and 21 (**Figure 4**).

Figure 5. Mean percentage change in objective daytime cough frequency at Day 22.

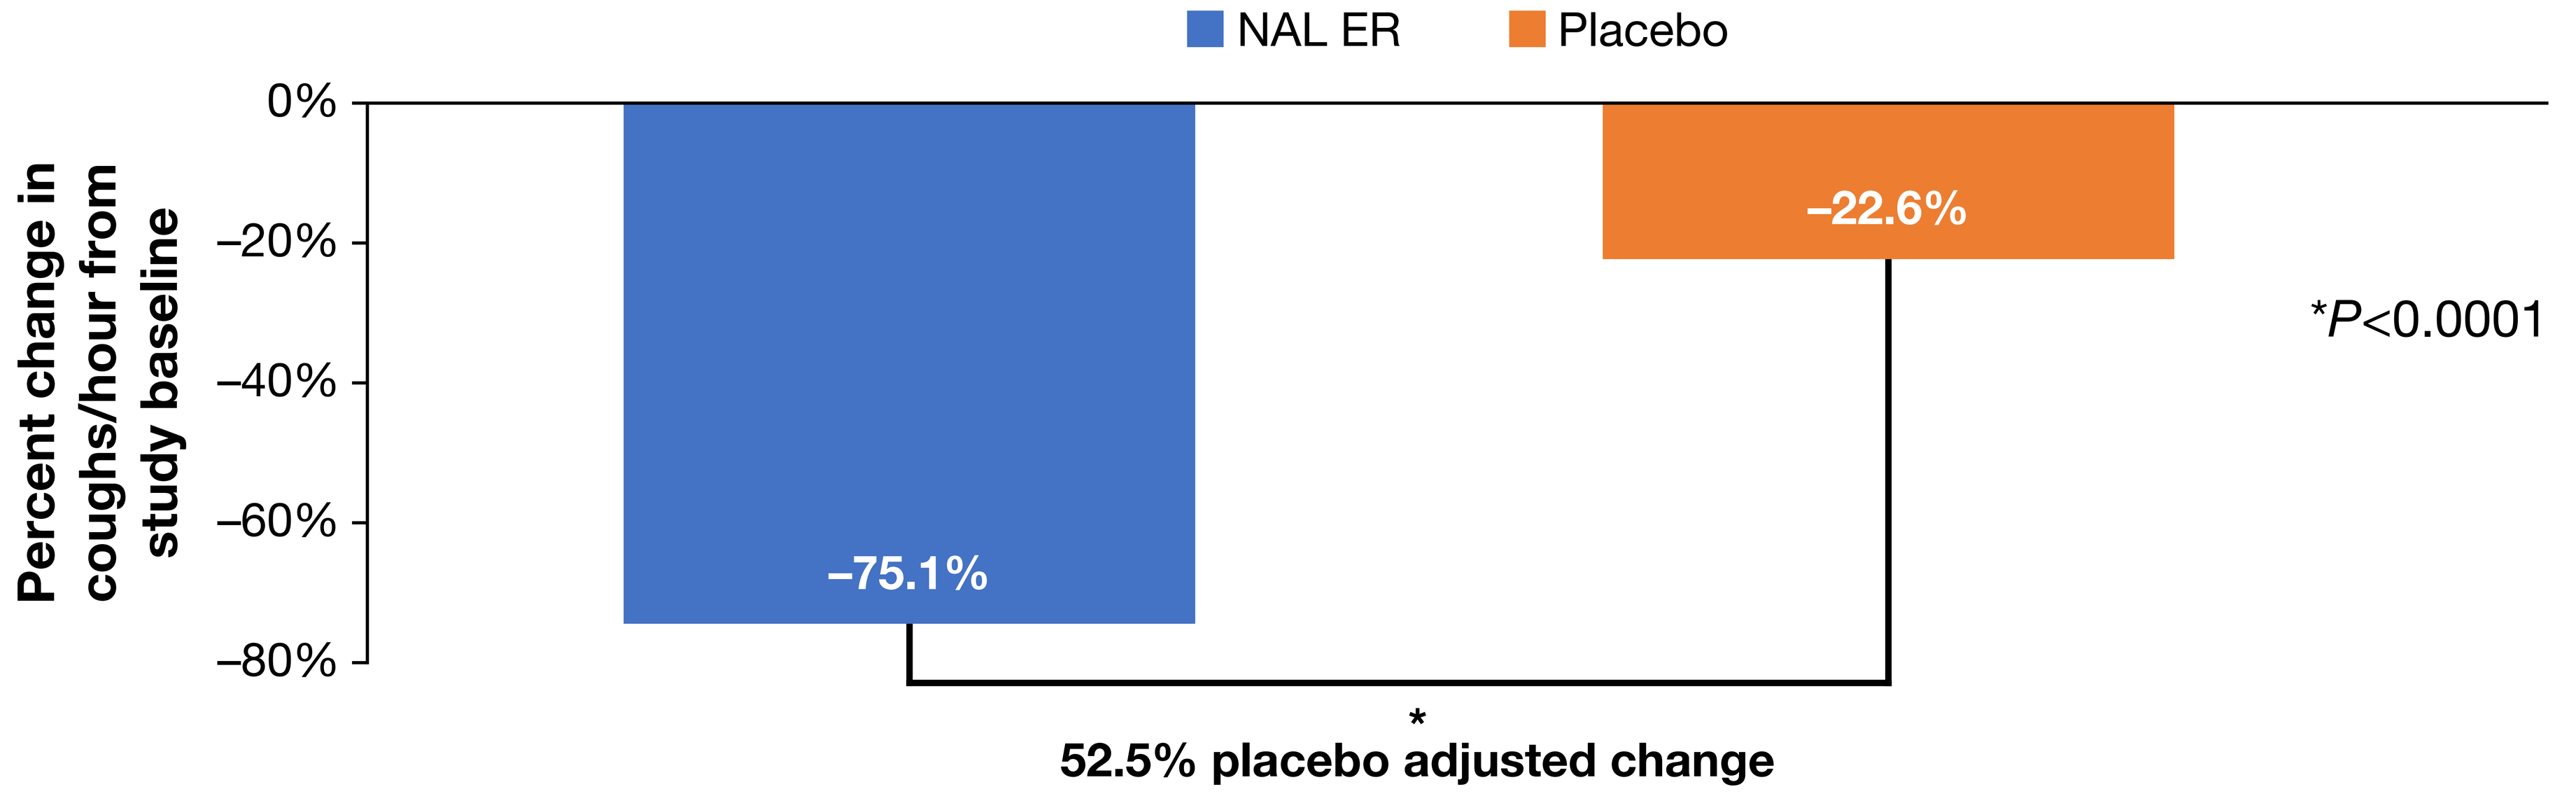
NAL ER
Placebo
Percent change in coughs/hour from study baseline
–75.1%
–22.6%
*P<0.0001
52.5% placebo adjusted change

NAL ER, nalbuphine extended release.

- The primary clinical-reported endpoint demonstrated a significant improvement of 52.5% placebo-adjusted change in daytime cough frequency for NAL ER at Day 22 (**Figure 5**).

CONCLUSIONS

- Overall EXACT and the E-RS breathlessness subdomain did not show significant separation between groups at Day 22 in these patients with IPF treated with nalbuphine ER.
- For patients treated with nalbuphine ER, the reduction in the patient reported E-RS cough score reflects the reduction in the primary endpoint of objective Daytime Cough Frequency at Day 22 vs. placebo.
 - A significant reduction in patient reported cough was seen from an early time point, with a dosage of only 54 mg, and was maintained over the duration of the study.
 - Patient reported E-RS cough subdomain is an easily repeatable PRO that reflects objective cough monitoring and could be a useful PRO in future IPF studies.

References: 1. Lee J, et al. Chest. 2022;162:603-13. 2. Bacci ED, et al. Respir Med. 2018;134:130-8. 3. Maher TM, et al. NEJM Evidence. 2023 (in press). doi:10.1056/EVIDoa2300083.

Acknowledgements: Research sponsored by Trevi Therapeutics. ClinicalTrials.gov Identifier: NCT04030026. Medical writing/editorial assistance was provided by Luke Ray, PhD, of Excerpta Medica, and was funded by Trevi Therapeutics, according to the Good Publication Practice guideline.

Disclosures: Dr Wijsenbeek: Research funding - The Netherlands Organization for Health Research and Development (ZonMw); The Dutch Pulmonary Fibrosis Patients Association (Longfonds); The Thorax Foundation, Erasmus MC; AstraZeneca-Daiichi; Hoffman la Roche; Boehringer Ingelheim. Speaker and/or consultancy fees - Boehringer Ingelheim; Bristol Myers Squibb; CSL Behring; Galapagos; Galecto; GSK; Hoffman la Roche; Horizon Therapeutics; Kinevant Sciences; Molecure; Nerre Therapeutics; Novartis; PureTech Health; Respivant; Thyron; Trevi Theraperutics; Vicore.

Presented at the 2023 European Respiratory Society (ERS) International Congress; 9–13 September, 2023; Milan, Italy.